EASL LIVER CANCER SUMMIT 2025
Paris, France
20–22 February

Final safety and efficacy results from the phase 1b/2a study of fostrox plus lenvatinib in second/third line advanced hepatocellular carcinoma progressed on immunotherapy

T.R. Jeffry Evans[1], Hong Jae Chon[2], Do Young Kim[3], Ho Yeong Lim[4], Teresa Macarulla[5], Carlos Gomez Martín[6], Victor Moreno[7], Min-Hee Ryu[8], Pia Baumann[9], Sujata Bhoi[9], Malene Jensen[9], Karin Tunblad[9], Hans Wallberg[9], Fredrik Öberg[9], Maria Reig[10], Jeong Heo[11]

[1]Beatson West of Scotland Cancer Center, Glasgow, United Kingdom, [2]CHA Bundang Medical Center, Seongnam, Korea, Rep. of South, [3]Severance Hospital, Yonsei University Health System, Seoul, Korea, Rep. of South, [4]Samsung Medical Center, Seoul, Korea, Rep.of South, [5]Hospital Vall d'Hebron, Barcelona, Spain, [6]Hospital Universitario 12 de Octubre, Madrid, Spain, [7]START Madrid-FJD, Madrid, Spain, [8]Asan Medical Center, Seoul, Korea, Rep. of South, [9]Medivir AB, Huddinge, Sweden, [10]Hospital Clinic de Barcelona, Barcelona, Spain, [11]Pusan National University Hospital, Busan, Korea, Rep. of South

MEDIVIR

Introduction

Fostrox is a liver targeted oral prodrug of troxacitabine that selectively inhibits DNA replication in liver tumors[1].

Fostrox monotherapy was investigated in a phase 1a/1b dose escalating and expansion study in advanced solid tumors with an encouraging safety, tolerability profile and preliminary anti-tumor activity in hepatocellular carcinoma (HCC)[2].

Based on these results, a phase 1b/2a study of fostrox in combination with lenvatinib (LEN) in second line in locally advanced unresectable or metastatic hepatocellular carcinoma (HCC) (NCT03781934) was initiated and end of treatment data are reported here.

Aim

The aim of the phase 1b/2a study was to establish a recommended phase 2 dose of fostrox in combination with LEN and to evaluate safety & tolerability together with preliminary anti-tumor activity data.

Method

Figure 1: Study design of phase 1b/2a fostrox + LEN in advanced HCC

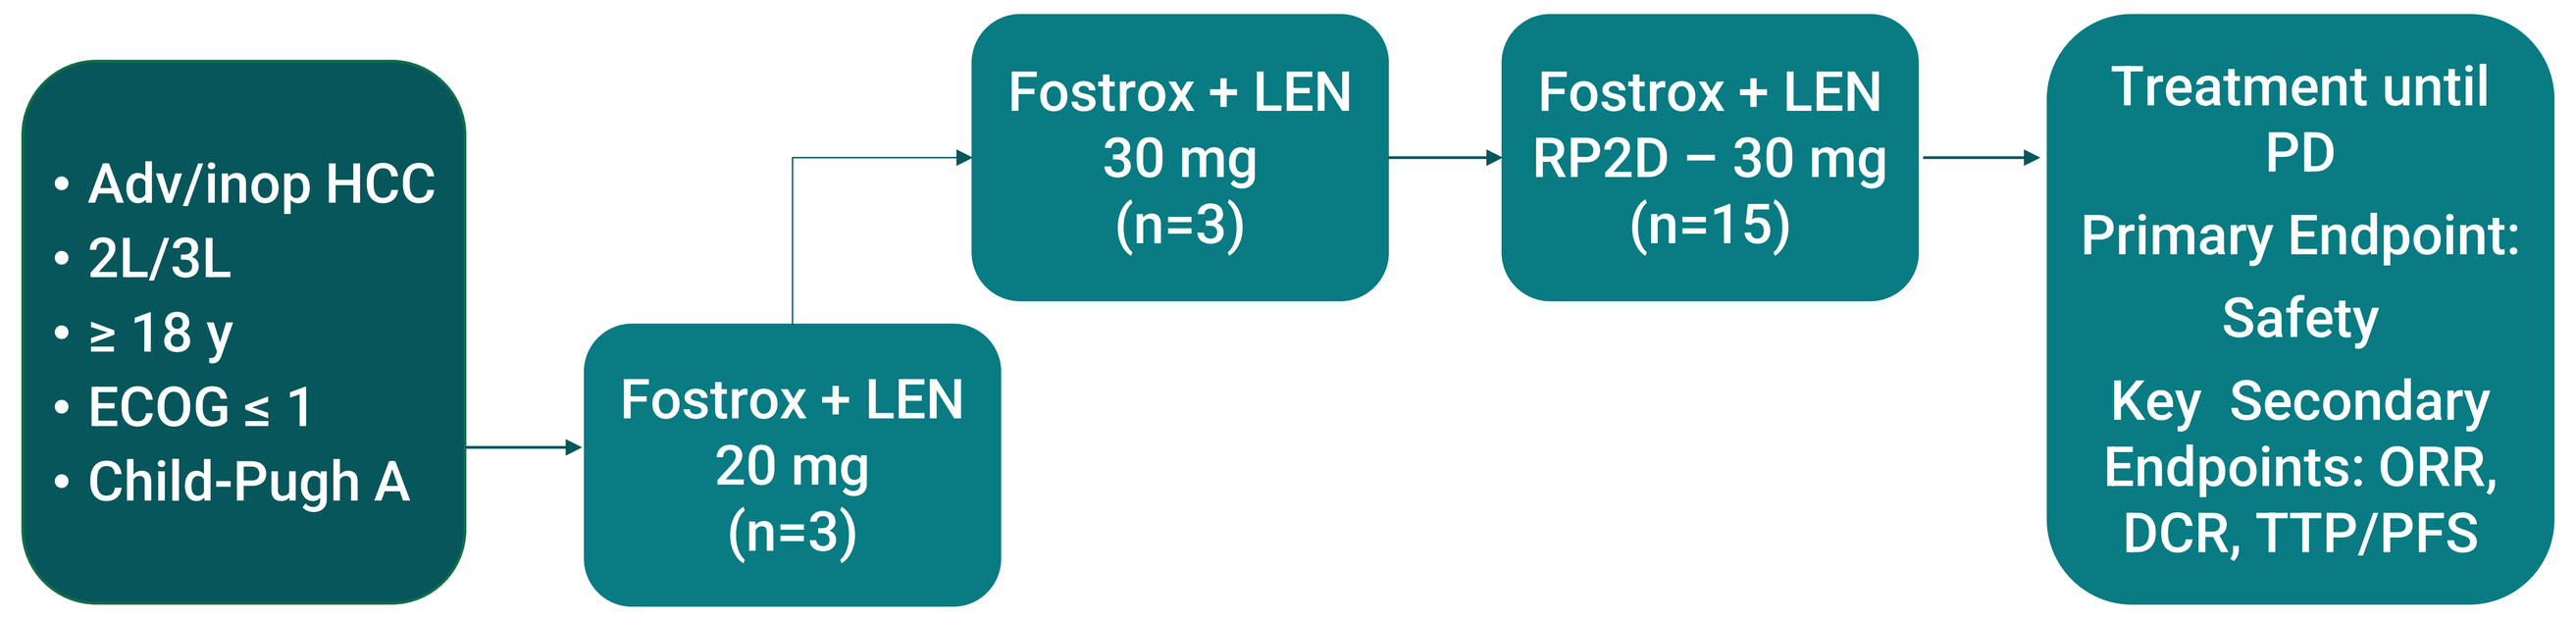
• Adv/inop HCC
• 2L/3L
• ≥ 18 y
• ECOG ≤ 1
• Child-Pugh A
Fostrox + LEN 20 mg (n=3)
Fostrox + LEN 30 mg (n=3)
Fostrox + LEN RP2D – 30 mg (n=15)
Treatment until PD
Primary Endpoint: Safety
Key Secondary Endpoints: ORR, DCR, TTP/PFS

Patients were enrolled at 15 sites in the UK, Spain and South Korea. Imaging assessments (CT & MRI) every 6 weeks.

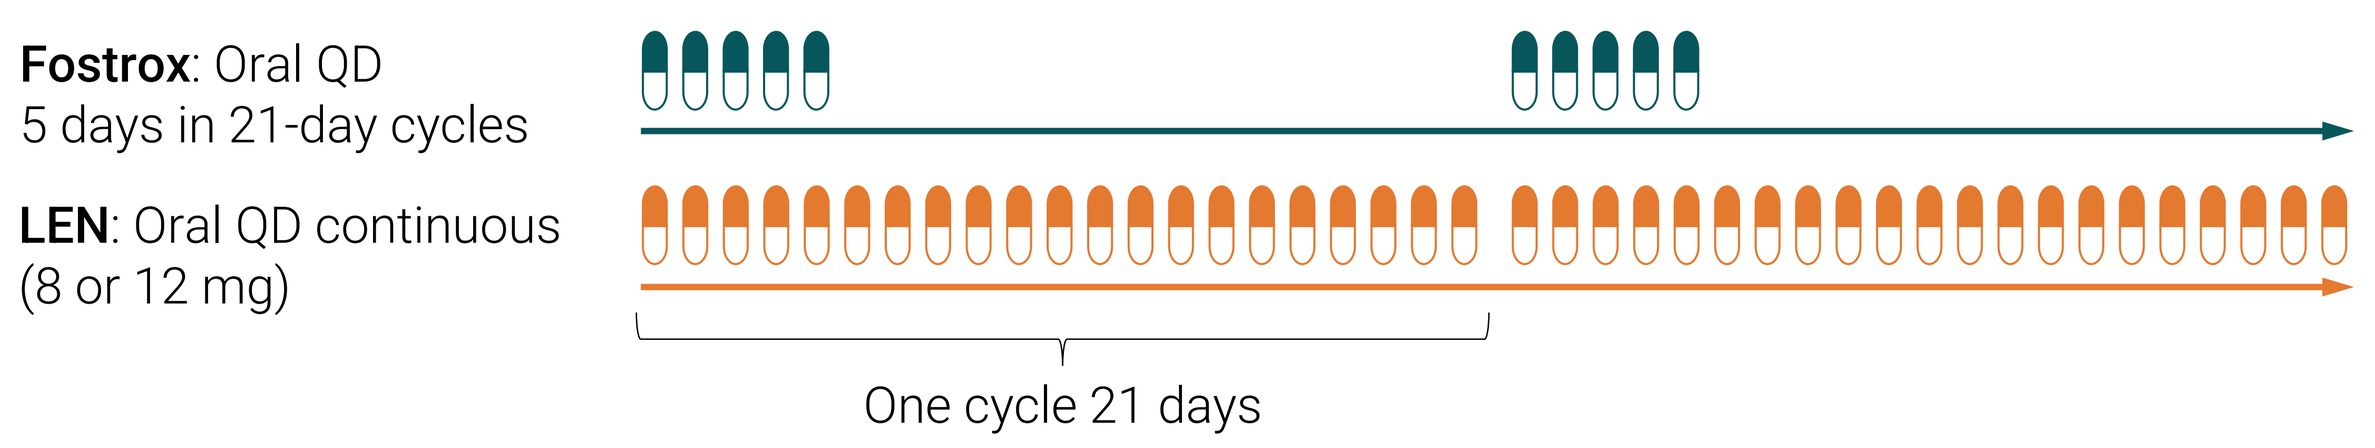
Fostrox: Oral QD
5 days in 21-day cycles
LEN: Oral QD continuous
(8 or 12 mg)
One cycle 21 days

Table 1: Patient characteristics

	N = 21
Median age (range)	63 yrs (42 - 82)
Gender, Female / Male (%)	24 / 76
ECOG Performance status 0/1 (%)	71 / 29
Child-Pugh A (%)	100
Viral/Non-viral (%)	76* / 24
Extra hepatic lesion(s) Y/N (%)	67 / 33
AFP ≥400 ng/mL at baseline Y/N (%)**	48 / 52
Region, Asia / Europe (%)	67 / 33
Prior treatment lines; 2nd line/3rd line (%)	81 /19
Prior atezolizumab/bevacizumab in 1L (%)	86
Prior local therapy (TACE, RFA etc)	70
PD on prior treatment (%)	100
Primary refractory on prior therapy (%)***	24
Starting dose fostrox, 20mg / 30mg (%)	14 / 86

*HepB-81% and HepC-19%; **AFP- NA for 1 pt; ***Active treatment ≤ 12 weeks. Data NA for 3 patients

Results

Safety and tolerability*

Table 2: Treatment emergent/related adverse events seen in >20% of patients

Adverse Events*	TEAE No of pts (%)			TRAE Grade ≥ 3* No of pts (%)	
	Grade 1/2	Grade 3	Grade 4	Fostrox	LEN
Hematologic AE					
Thrombocytopenia	13 (62)	5 (24)	2 (10)	5 (24)	6 (29)
Neutropenia (no febrile)	9 (43)	8 (38)	2 (10)	8 (38)	6 (29)
Anaemia	6 (29)	3 (14)		3 (14)	3 (14)
Leukocyte decrease	6 (29)	2 (10)		2 (10)	2 (10)
Other AE					
Hypothyroidism	11 (52)				
Diarrhea	11 (52)	1 (5)			1 (5)
Hand-foot syndrome	10 (48)	1 (5)			1 (5)
Fatigue	9 (43)				
Asthenia	2 (10)	2 (10)		1 (5)	2 (10)
Anorexia	8 (38)				
Proteinuria	7 (33)	2 (10)			1 (5)
Hypertension	8 (38)	3 (14)			2 (10)
Cough	5 (24)				
Pruritus	5 (24)				

* There was no grade 5 fostrox related AE and one grade 5 LEN related death (renal failure)

- With a median follow-up of 10.5 months (range 1.2 – 27.8 months) the combination of fostrox + LEN was shown to be safe and well tolerated with no unexpected new adverse events reported.
- Most common adverse events were laboratory test results with 33 events (out of 880 blood tests) of grade ≥ 3 neutropenia and thrombocytopenia with clinical relevance (fostrox: dose-delay, reduction or discontinuation) in 11 of these 33 occasions.
- No febrile neutropenia or low platelet count with bleeding was reported
- SAEs were reported in 8 patients with 17 events in total
 - No events were related to fostrox
 - 8 events in 6 patients were possibly related/related to LEN (asthenia, ischemic stroke, renal failure, hepatic encephalopathy, diarrhea)
- Fostrox dose reduction due to AE was done in 29% and discontinuation in 5%
- LEN dose reduction due to AE was done in 52% and discontinuation in 10%

*Data cut Nov 30 2024

Clinical efficacy* (investigator review, RECISTv1.1)

Figure 2. Time to progression (TTP)

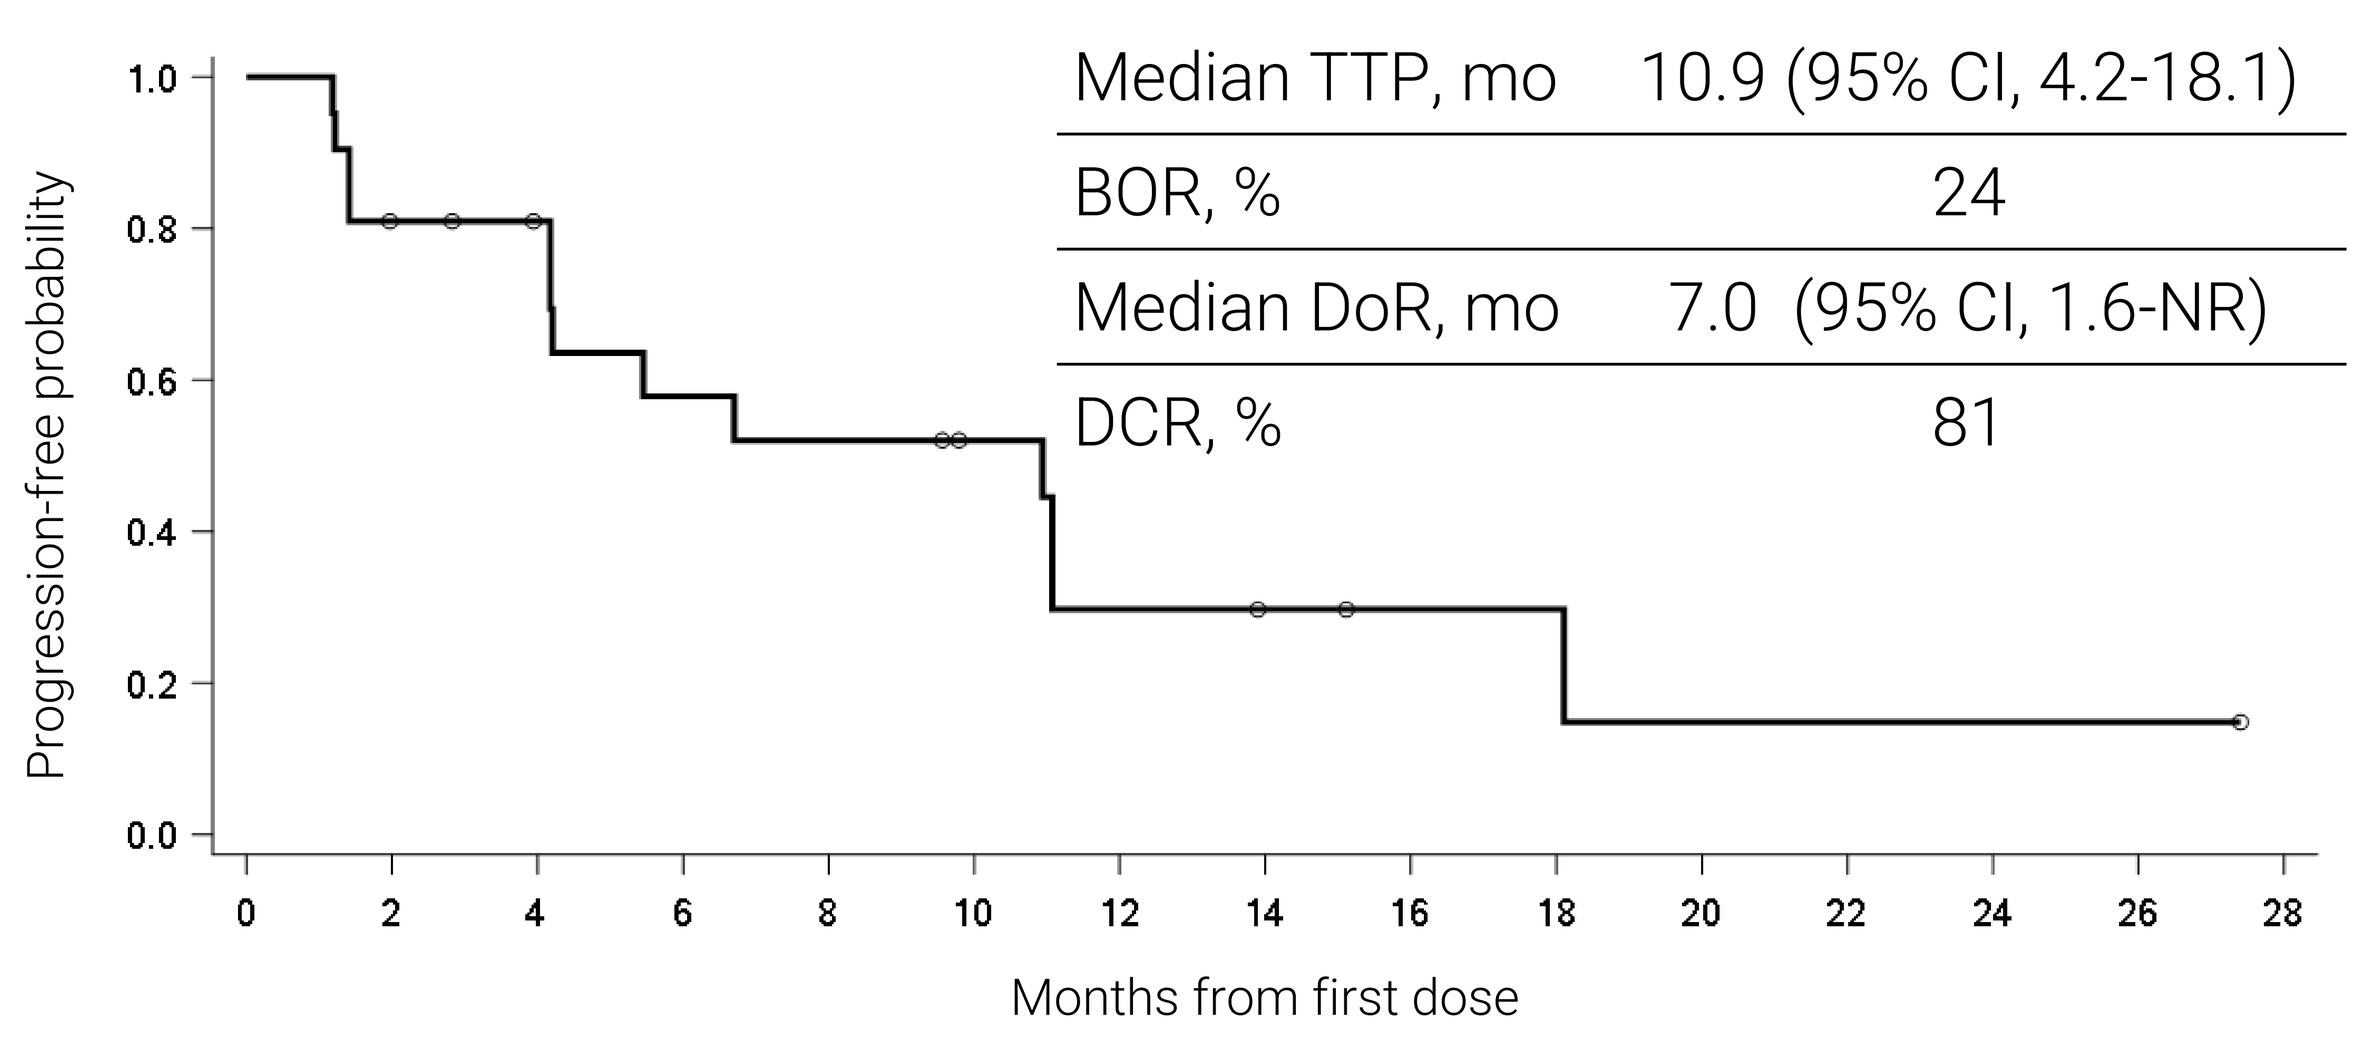
Median TTP, mo 10.9 (95% CI, 4.2-18.1)
BOR, % 24
Median DoR, mo 7.0 (95% CI, 1.6-NR)
DCR, % 81
Progression-free probability
Months from first dose

Figure 3. Overall survival (OS)

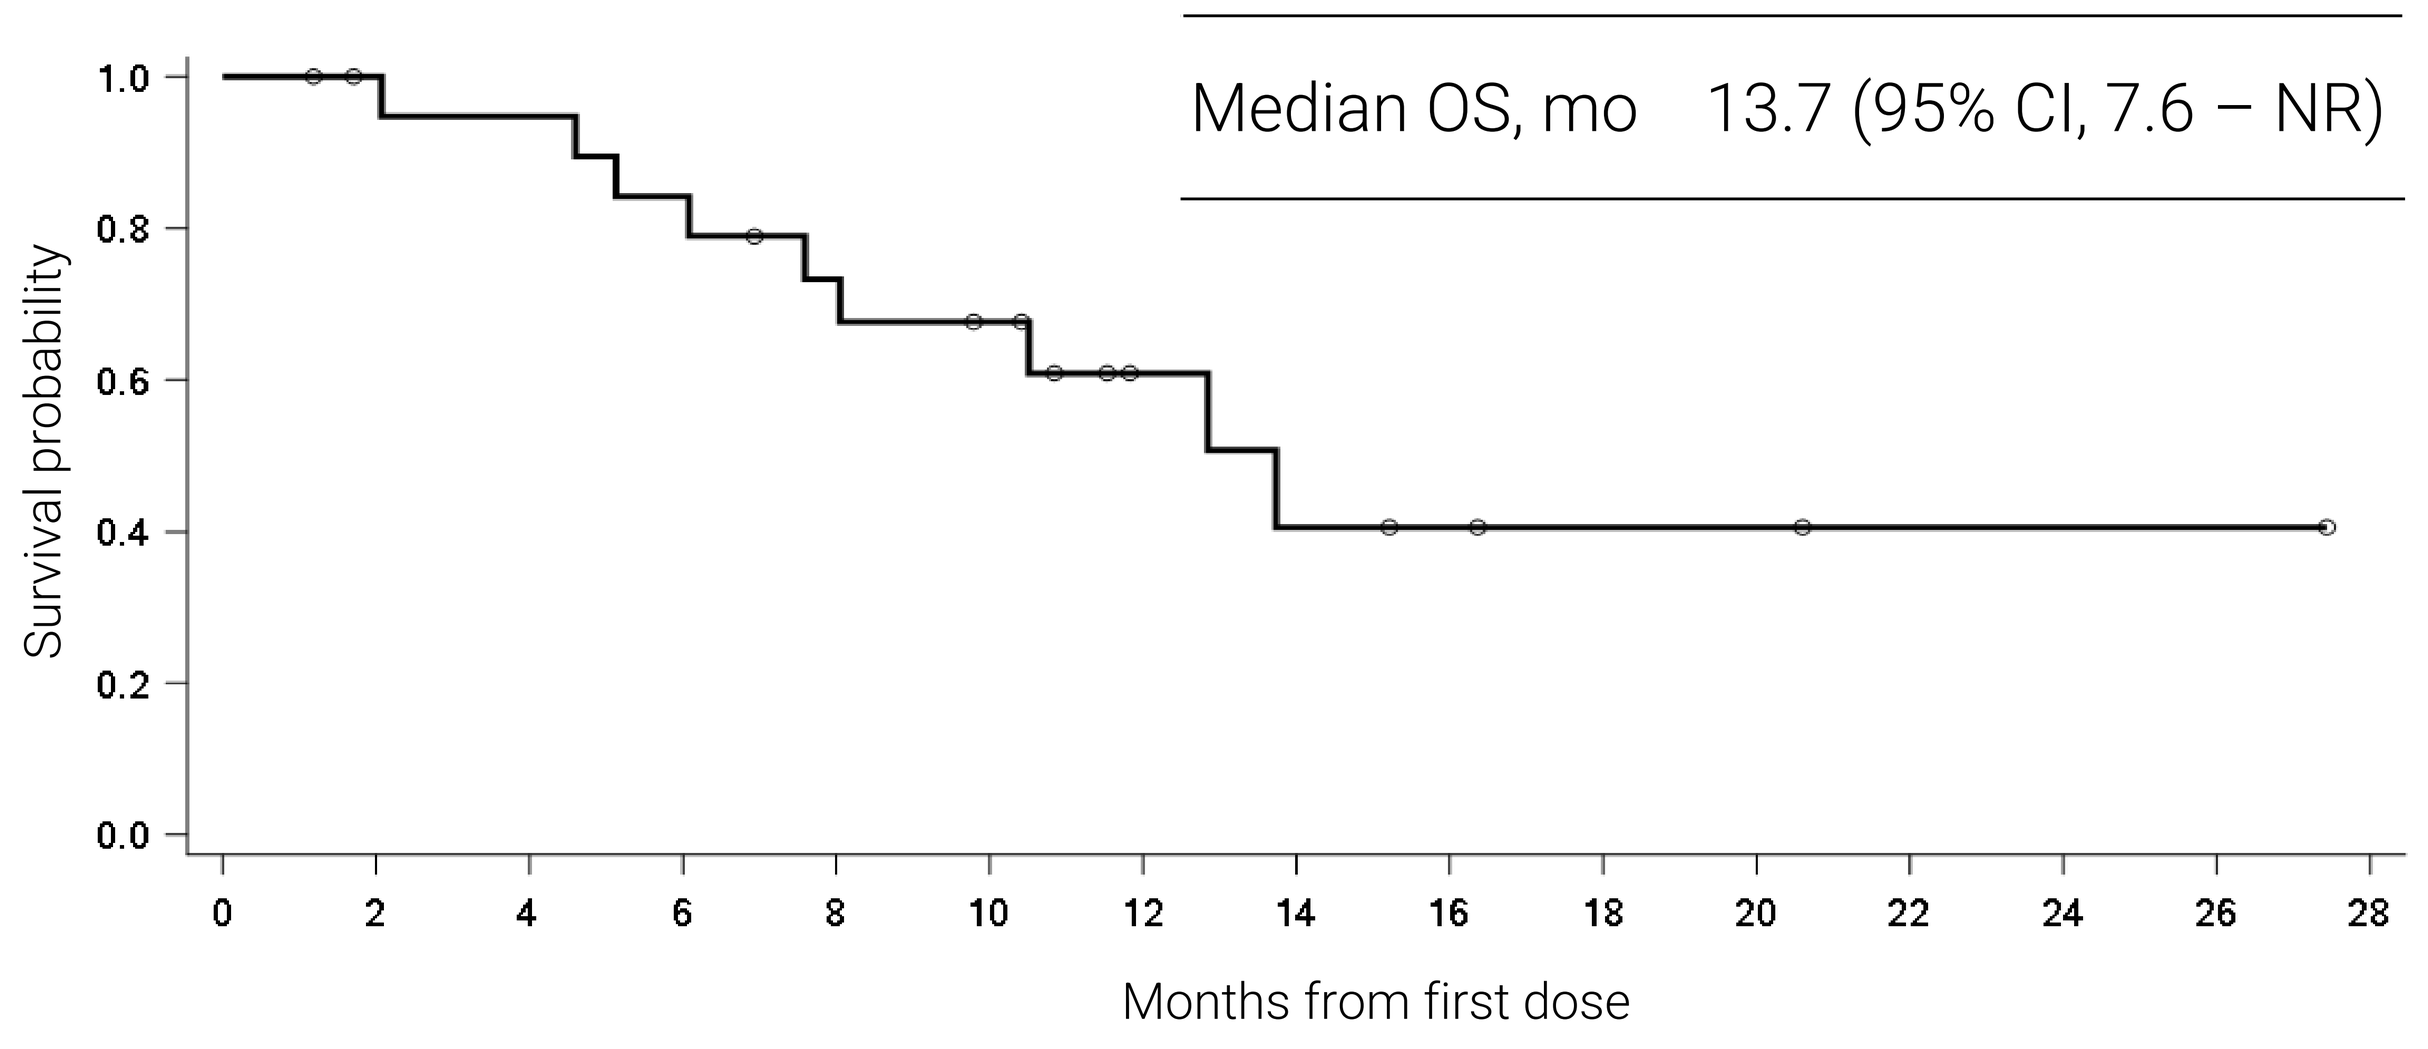
Median OS, mo 13.7 (95% CI, 7.6 – NR)
Survival probability
Months from first dose

Figure 4. Percentage change in total target lesion size over time

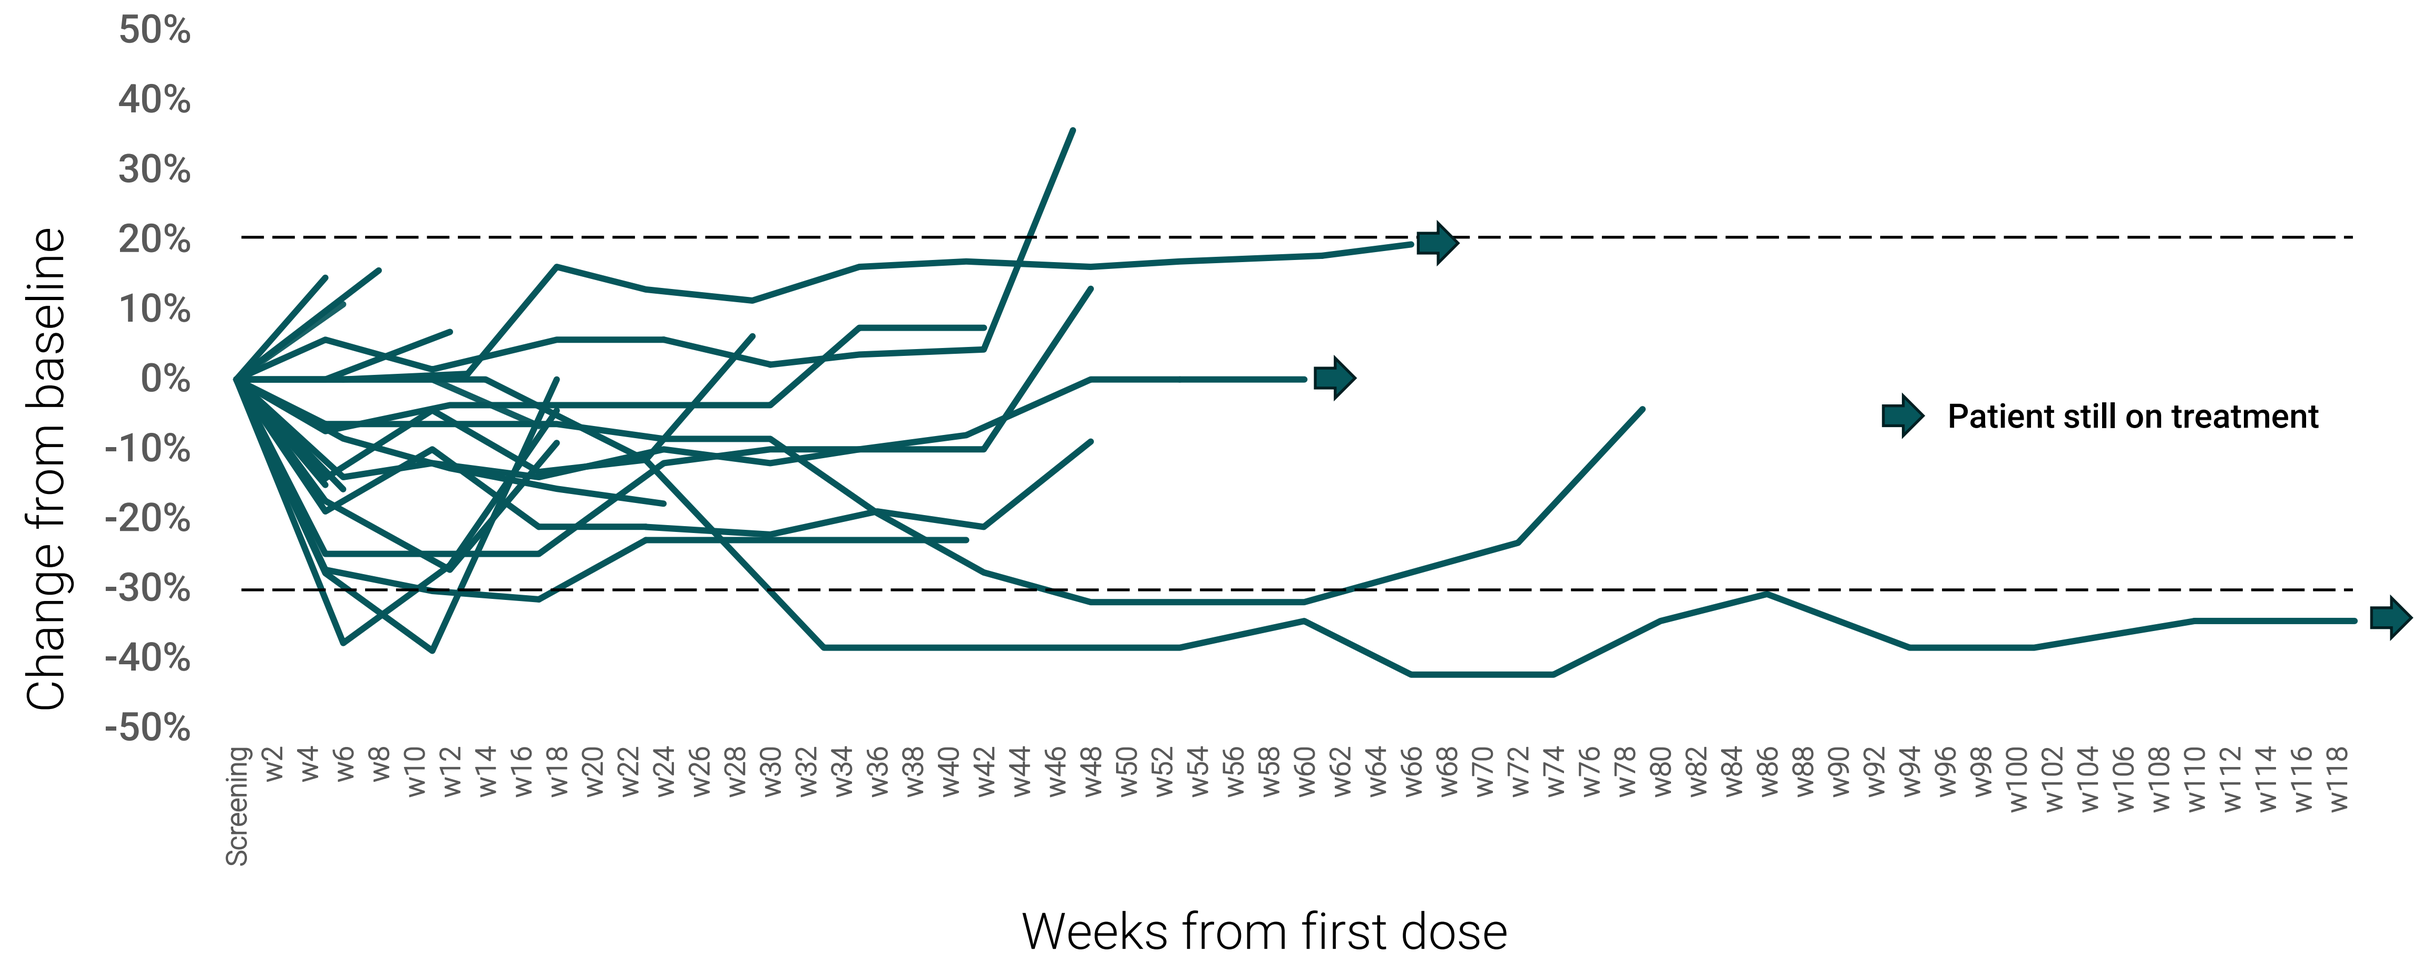
Change from baseline
Patient still on treatment
Weeks from first dose

Target lesion tumor reduction was seen in >75% of patients and median duration of clinical benefit (CR, PR or SD>24 weeks[2]) was 11.3 months.

Figure 5. Best % change in target lesion size related to fostrox dose reduction

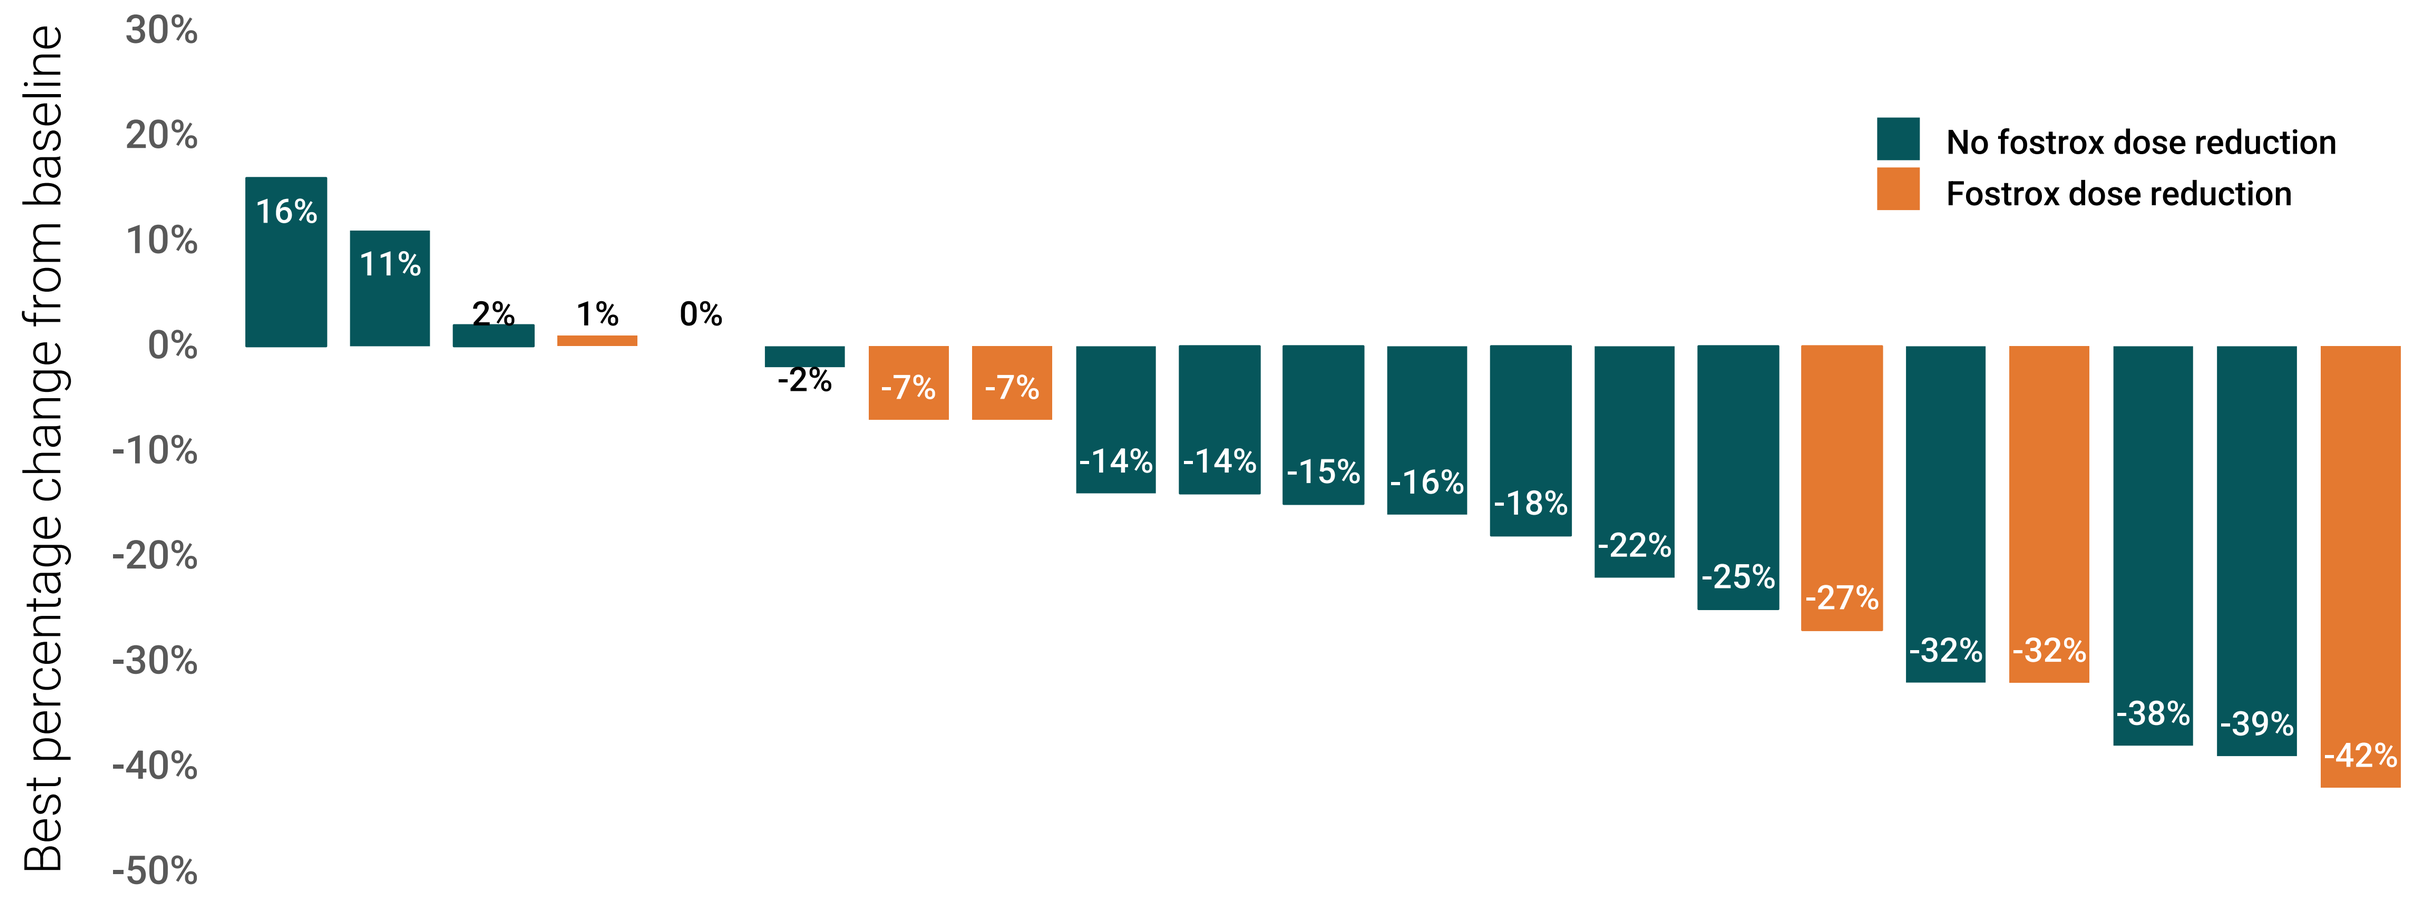
No fostrox dose reduction
Fostrox dose reduction
Best percentage change from baseline
16% 11% 2% 1% 0% -2% -7% -7% -14% -14% -15% -16% -18% -22% -25% -27% -32% -32% -38% -39% -42%

Fostrox dose reduction, required in 29% of patients, didn't impact response to treatment

Figure 6. Percentage change in AFP over time and best overall response

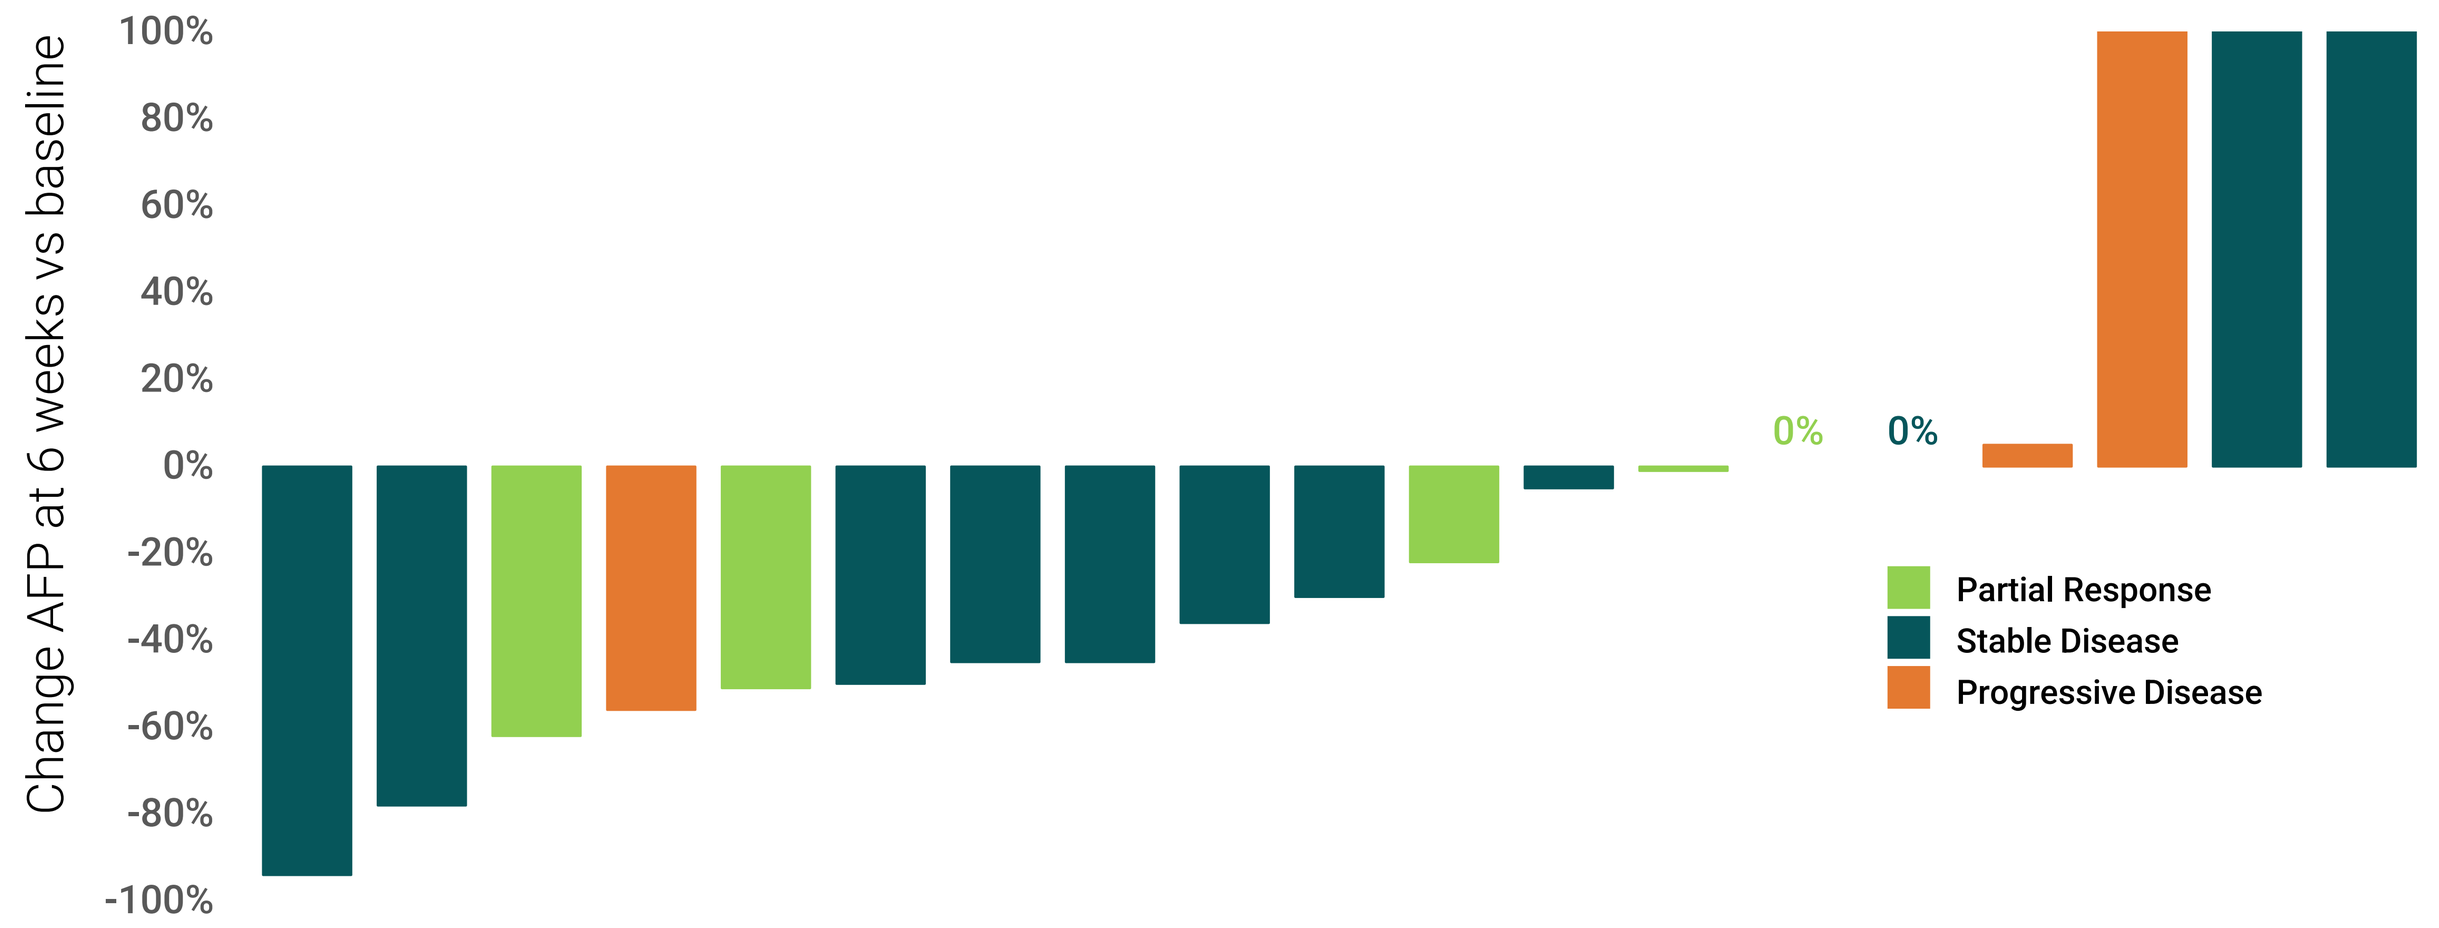
Change AFP at 6 weeks vs baseline
0% 0%
Partial Response
Stable Disease
Progressive Disease

Reduced or stable AFP levels from baseline to 6 weeks were seen in 79% of patients and was associated with imaging response (RECISTv1.1).

Figure 7. Best % change in target lesion size related to AFP at baseline

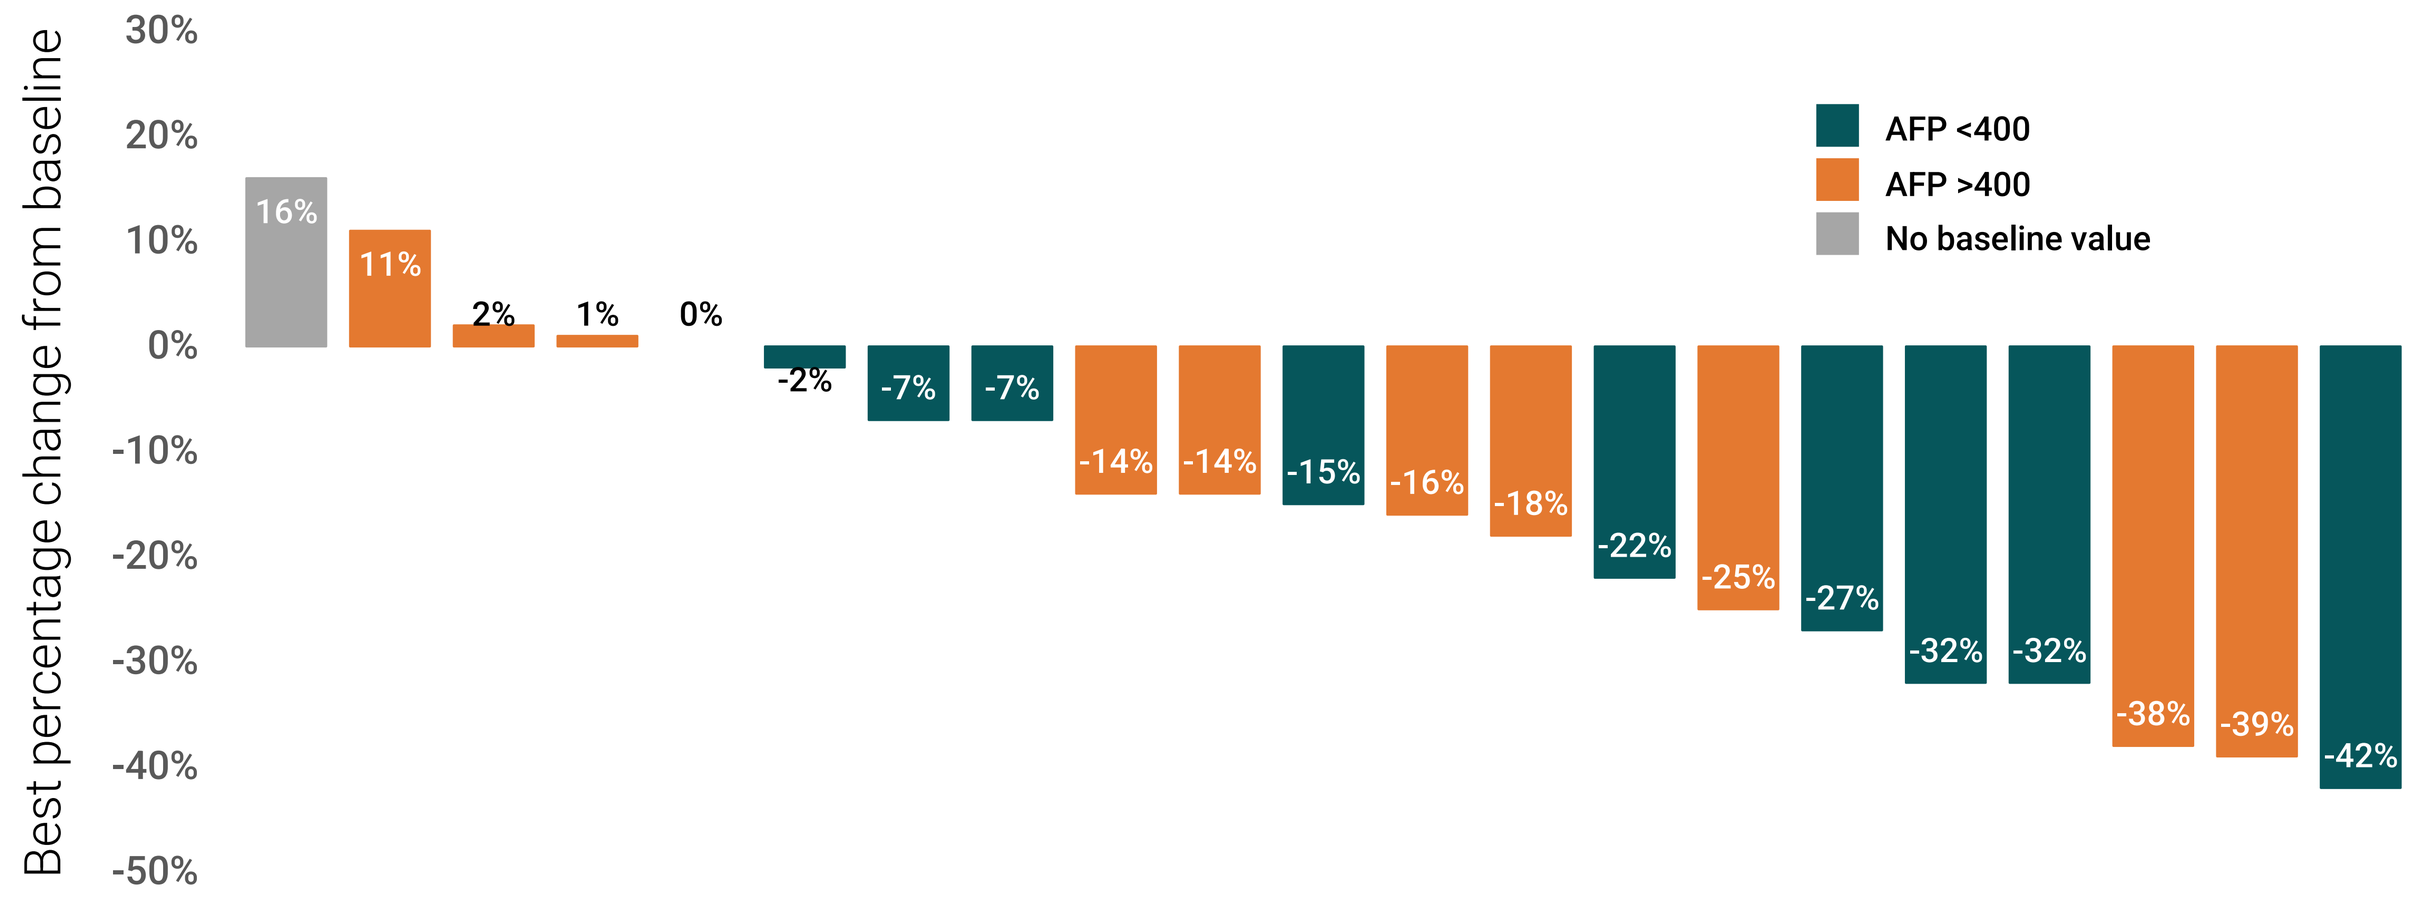
AFP <400
AFP >400
No baseline value
Best percentage change from baseline
16% 11% 2% 1% 0% -2% -7% -7% -14% -14% -15% -16% -18% -22% -25% -27% -32% -32% -38% -39% -42%

Target lesion size reduction was seen independent of AFP level at baseline

Conclusions

- **Fostrox + LEN was safe and well tolerated using the RP2D of fostrox 30 mg QD orally for 5 days in a 21-day cycle in combination with standard dose of LEN**
- **Encouraging efficacy was seen with a best objective response, median time to progression and overall survival of 24%, 10.9 months and 13.7 months, respectively**
- **The majority of patients had a reduction in AFP levels at 6 weeks which correlated with imaging response, and overall target lesion reduction was independent of baseline AFP levels**
- **Based on the promising result from this study, a randomized phase 2b study with fostrox + LEN versus placebo + LEN in second line advanced HCC is planned to enroll patients in Europe, Asia and in North America**

Acknowledgement

Thanks to all the investigators, research teams, and clinical staff in Korea, Spain and the UK, whose dedication and expertise made this study possible. We also sincerely thank the patients and their families for participating and contribution to advancing medical knowledge in liver cancer.

Contact information

j.evans@crukscotlandinstitute.ac.uk
pia.baumann@medivir.com

Reference

1. Bethell, R. et al 2017, EASL International Liver Congress, Poster SAT-123 (www.medivir.com/media/1196/liver-targeting-with-the-novel-nucleotide-prodrug-miv-818.pdf)
2. J Hepatocell Carcinoma,. 2024 Oct 24:11:2033-2047
3. Am J Cancer Res 2021;11(4):1121-1131

Abbreviations

Heptatocellular carcinoma (HCC), recommended phase 2 dose (RP2D), progressive disease (PD), best objective response (BOR), duration of response (DoR), disease control rate (DCR), time to progression (TTP), duration of clinical benefit (DoCB), Eastern Cooperative Oncology Group Performance Status (ECOG PS), Response evaluation criteria in solid tumors (RECIST 1.1), Computerized tomography (CT), Magnetic Resonance Imaging (MRI), Trans arterial chemoembolisation (TACE), radio frequency ablation (RFA), Common Terminology Criteria for Adverse Events (CTCAE), Immunotherapy (IO), target lesion (TL), treatment emergent adverse event (TEAE), treatment related adverse event (TRAE), lenvatinib (LEN), Kaplan-Meier (K-M), serious adverse event (SAE), quaque die/once daily (QD), alfa-feto protein (AFP), overall survival (OS)